Administration of SP-101 and doxorubicin results in robust and durable hCFTRΔR transgene expression in the airways of ferrets

POSTER 621
(also see Poster 672)

Katherine Excoffon[1], Mark Smith[1], Lillian Falese[1], Robert Schulingkamp[1], Shen Lin[1], Poornima Narayan[1], Madhu Mahankali[1], Matt Glatfelter[1], Eric Yuen[1] and Roland Kolbeck[1]

[1]Spirovant Sciences, Inc., Philadelphia, PA

SP-101 – A novel, inhaled AAV-based gene therapy to treat CF

DESIGN FEATURES

- AAV capsid selected for tropism to the apical surface of human airway epithelia (HAE)[1]
- hCFTRΔR minigene with regulatory elements[2,3]

MECHANISM OF ACTION

- Efficient apical entry
- Enhanced SP-101 translocation to the nucleus by administration of doxorubicin (Dox)[4,5]
- Increased CFTR expression

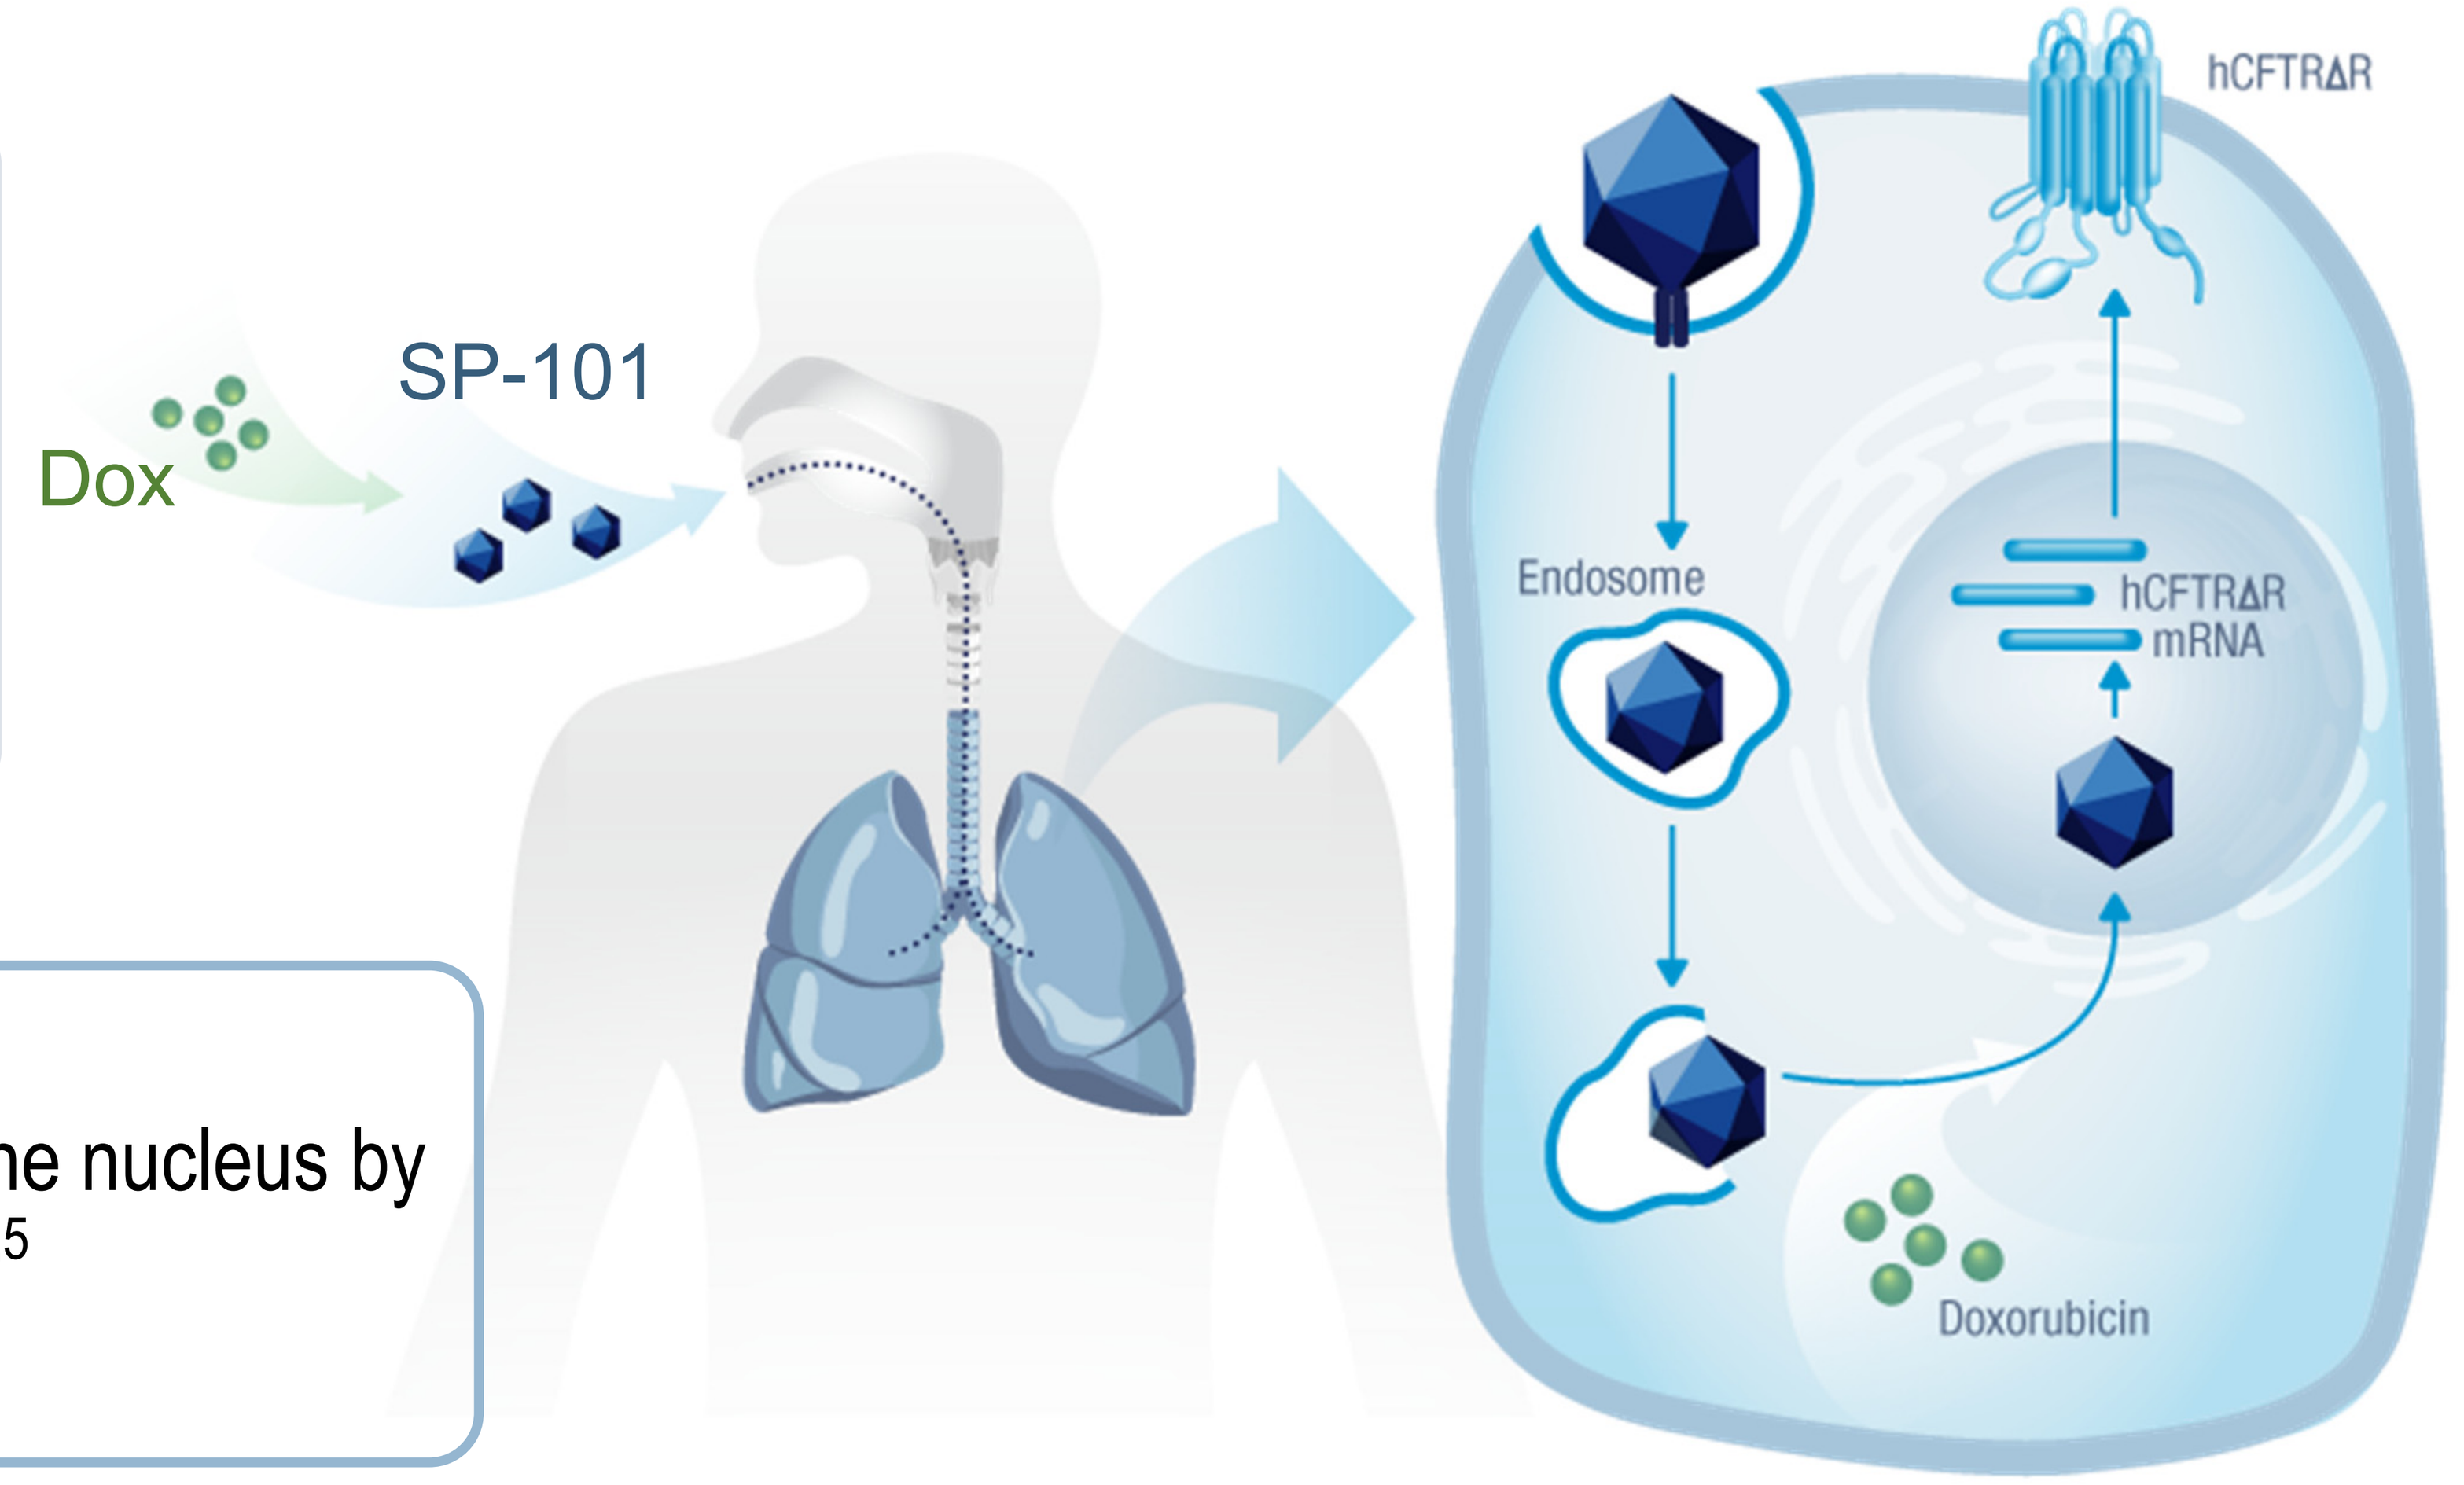
Dox
SP-101
hCFTRΔR
Endosome
hCFTRΔR mRNA
Doxorubicin

Ferret as a model to evaluate inhaled SP-101

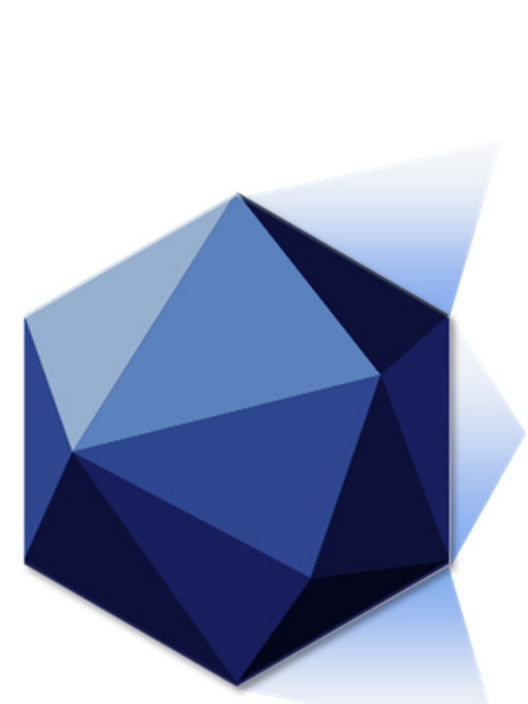

SP-101 capsid is tropic to ferret airway cells[6]

CF ferret model recapitulates human CF lung pathology[7]

Ability to administer via inhalation

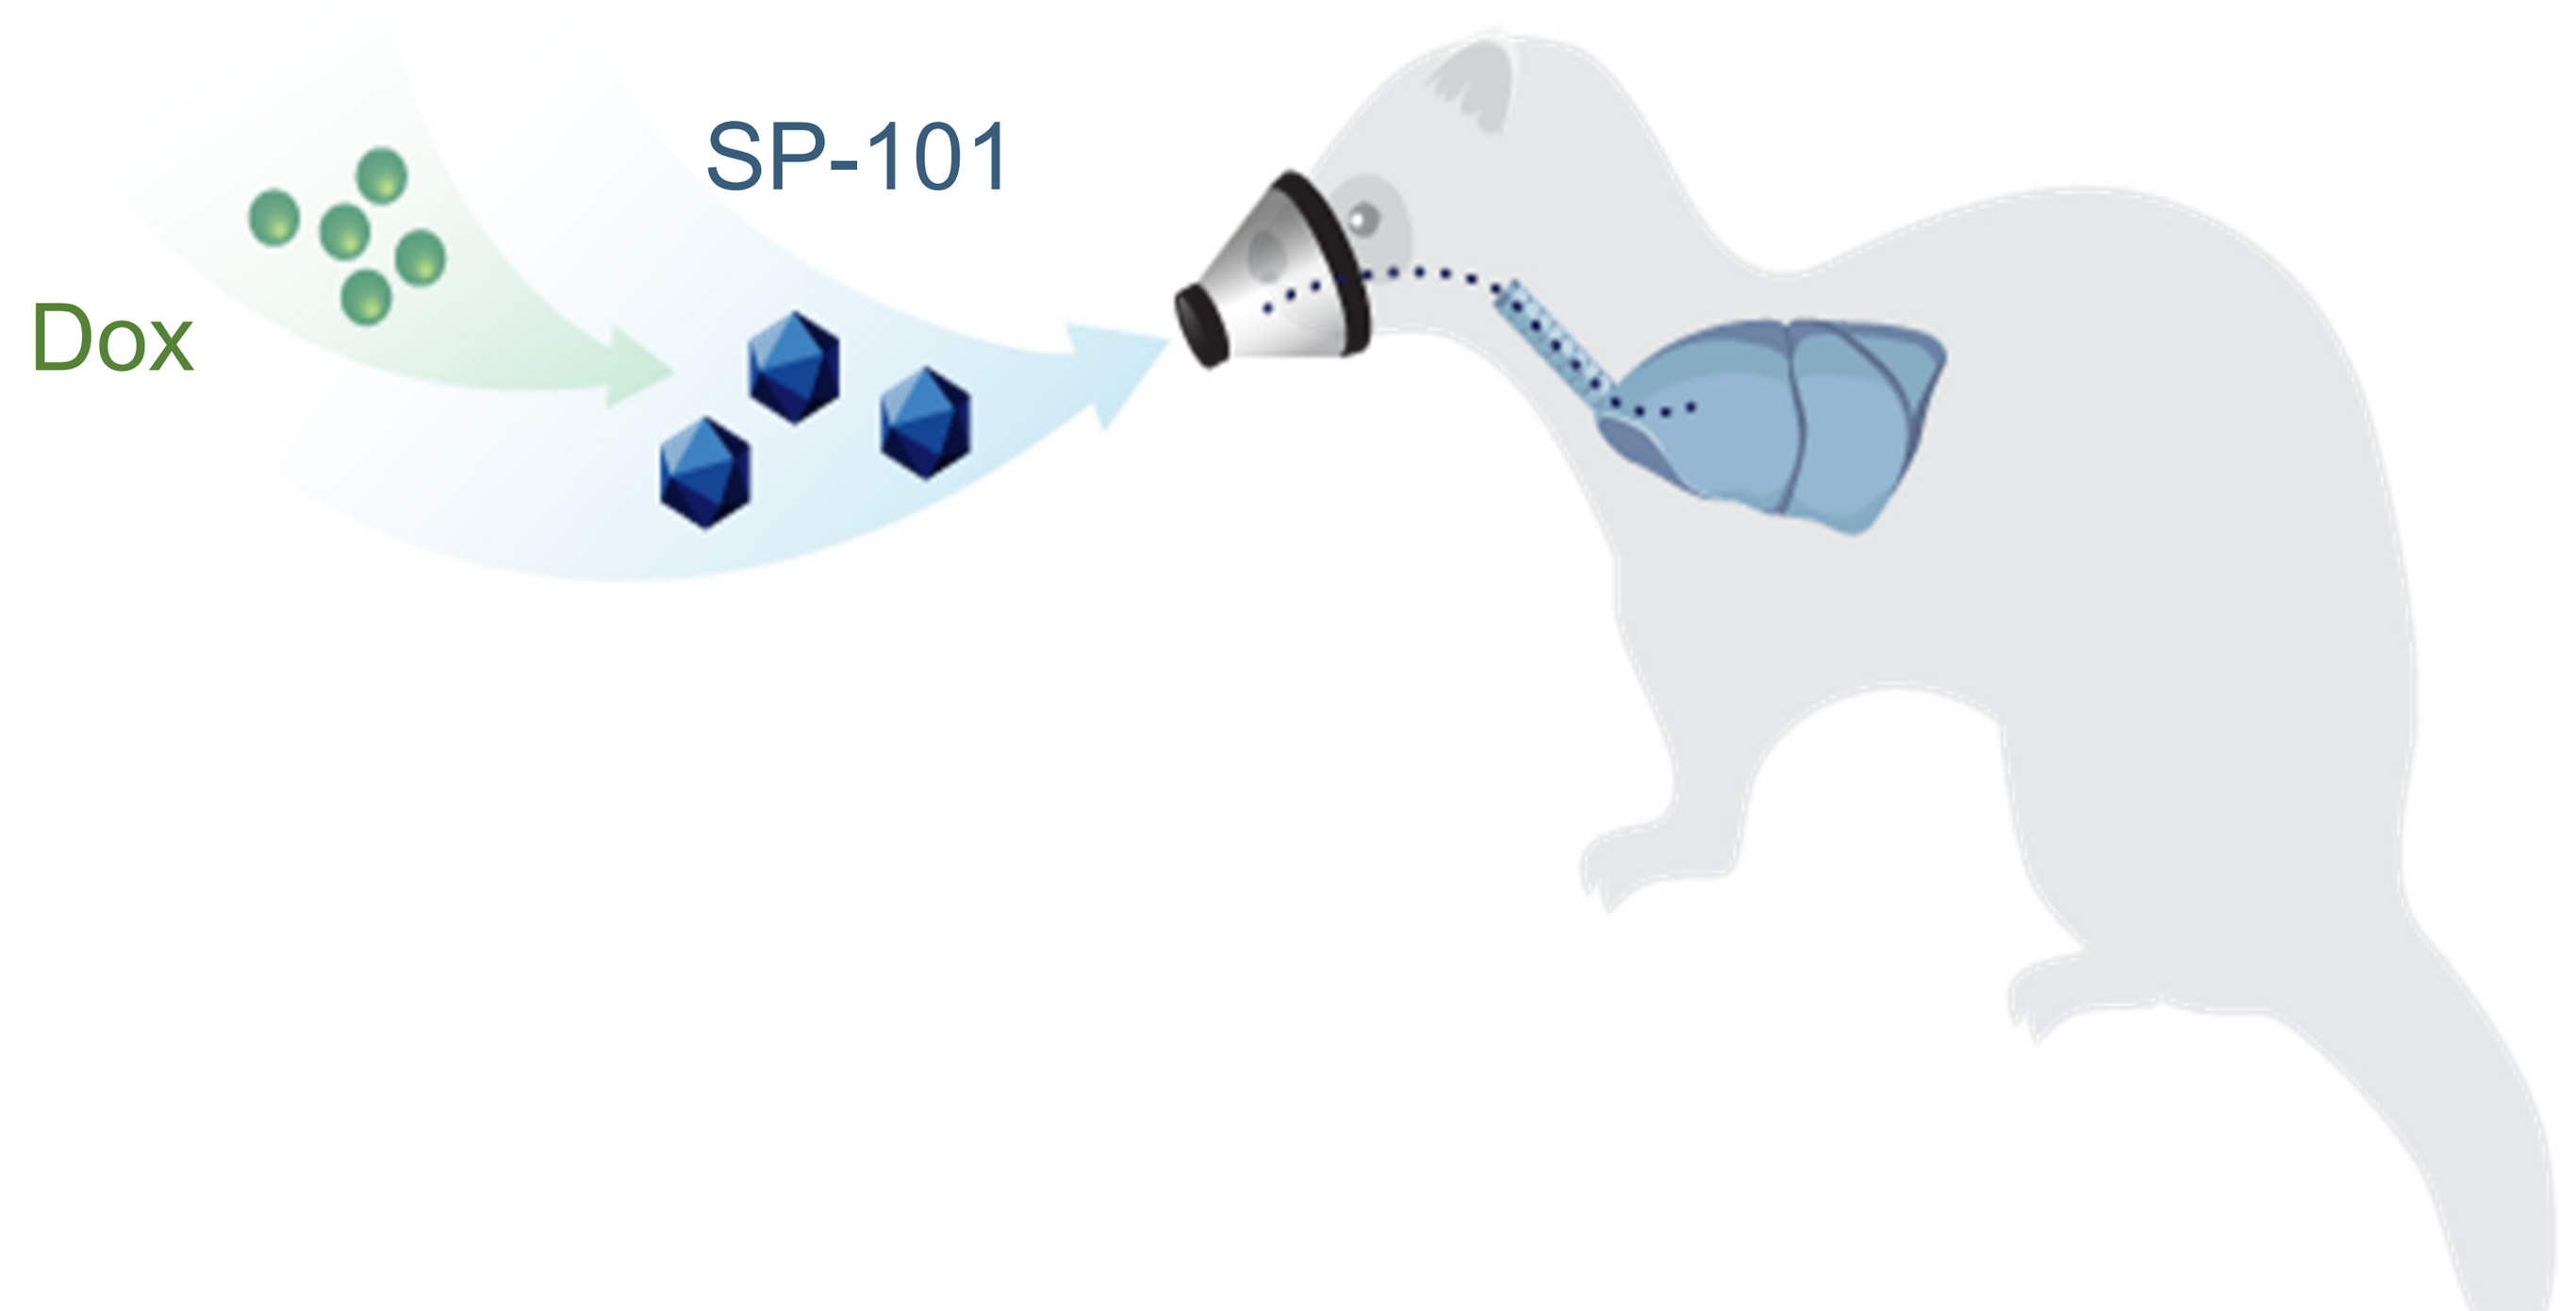
Dox
SP-101

Methods: Non-CF and CF ferrets were exposed to nebulized SP-101 or placebo, followed by doxorubicin (dox) or placebo on Day 1. Animals were necropsied 2 or 12 weeks post-exposure and tissues harvested for *in situ* hybridization (ISH) or determination of hCFTRΔR mRNA copy count.
ISH: Sections from formalin-fixed, paraffin-embedded lung were evaluated by RNAScope, using zz-probes designed to the sense strand unique regions of the SP-101 vector genome.
hCFTRΔR mRNA copies: RNA was isolated, using a DNase procedure to ensure removal of vector genomes, from ~25 mg samples taken from 8-9 different regions of the airway (tracheal, bronchial, lobe). qPCR +/- reverse transcriptase was performed with primers and a probe for a unique region of the *hCFTRΔR* mRNA. No signal was observed in the absence of reverse transcriptase indicating the complete removal of vector genomes (data not shown). Data are shown as box and whisker plots around the median value (*hCFTRΔR* copy count normalized to 500ng total RNA).

SP-101 is tropic to the ferret airway

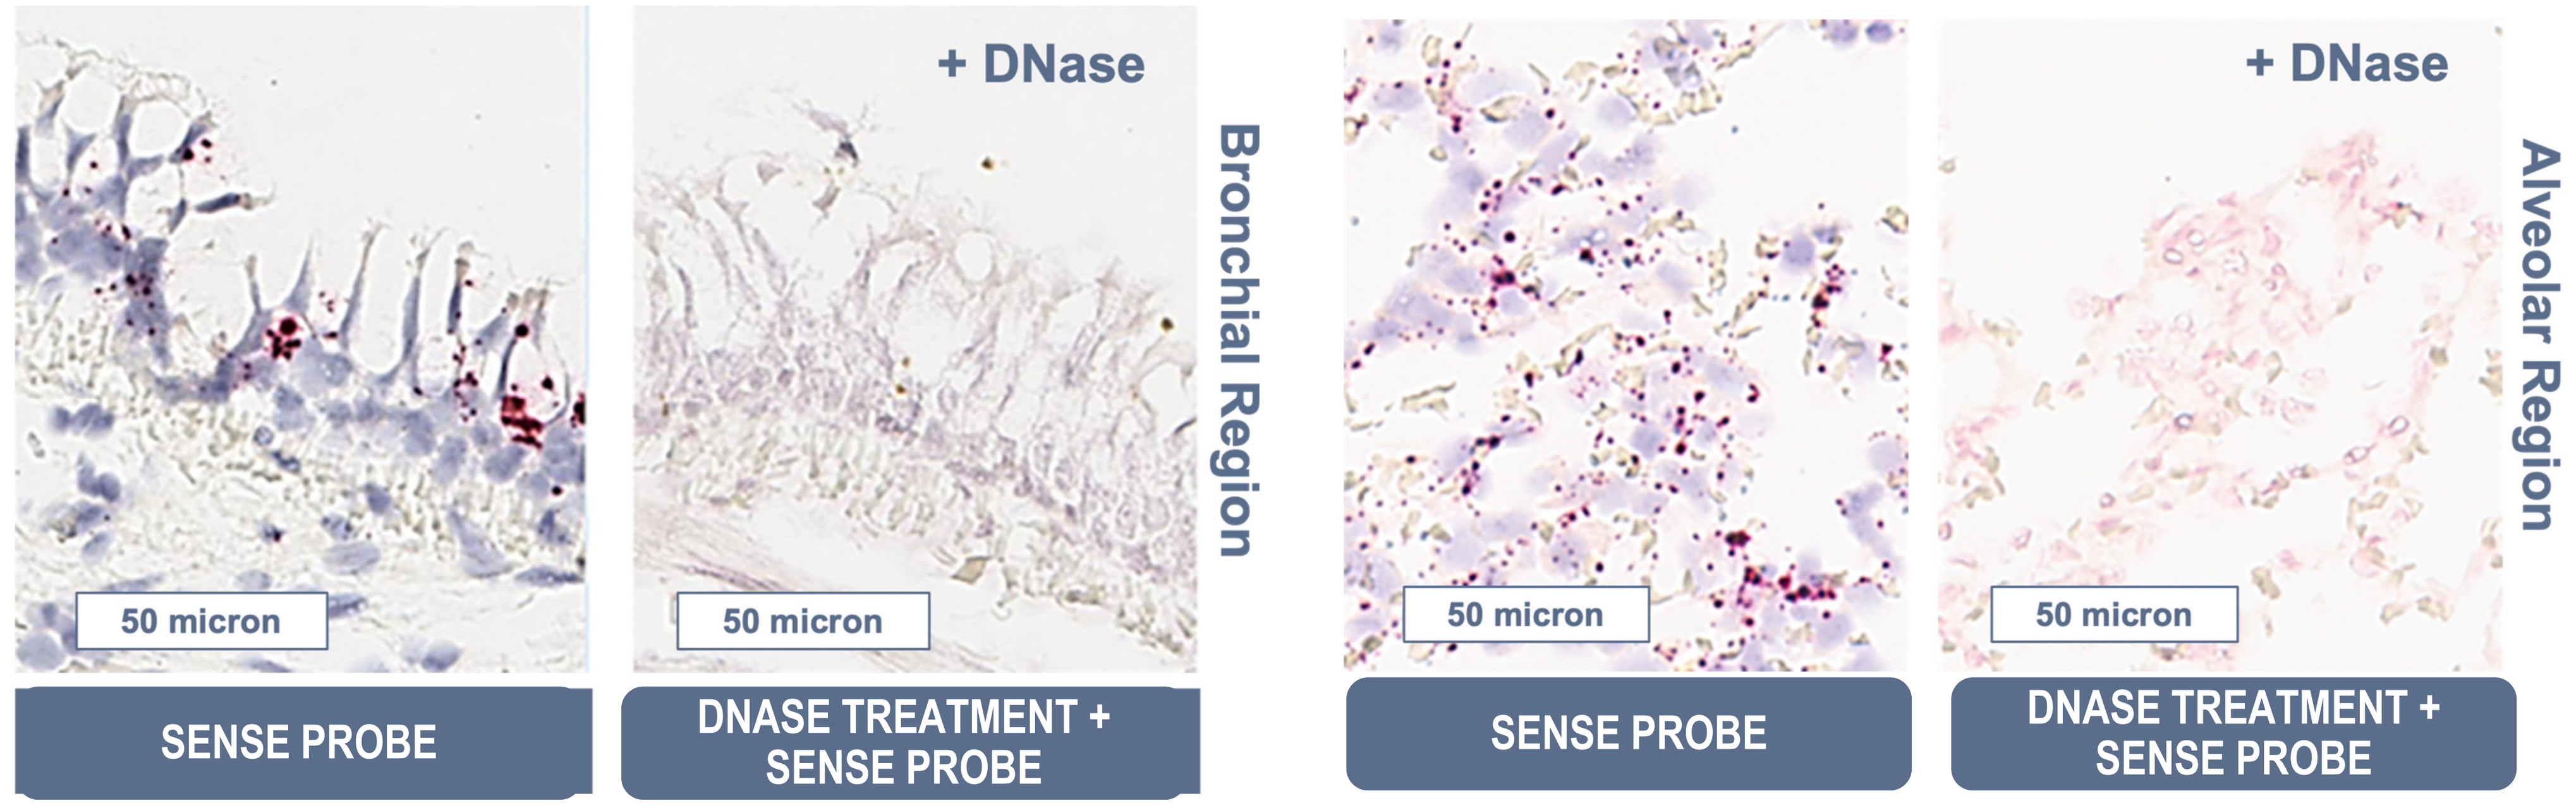
+ DNase
Bronchial Region
50 micron
50 micron
SENSE PROBE
DNASE TREATMENT + SENSE PROBE
+ DNase
Alveolar Region
50 micron
50 micron
SENSE PROBE
DNASE TREATMENT + SENSE PROBE

SP-101 vector genomes are abundant in many regions of non-CF ferret lungs. SP-101 vector genomes (red dots) were detected in multiple cells whereas pretreatment with DNase did not show staining indicating the specificity of staining.

hCFTRΔR mRNA expression is enhanced by doxorubicin, dose responsive, durable, and similar in both CF and wild-type ferrets

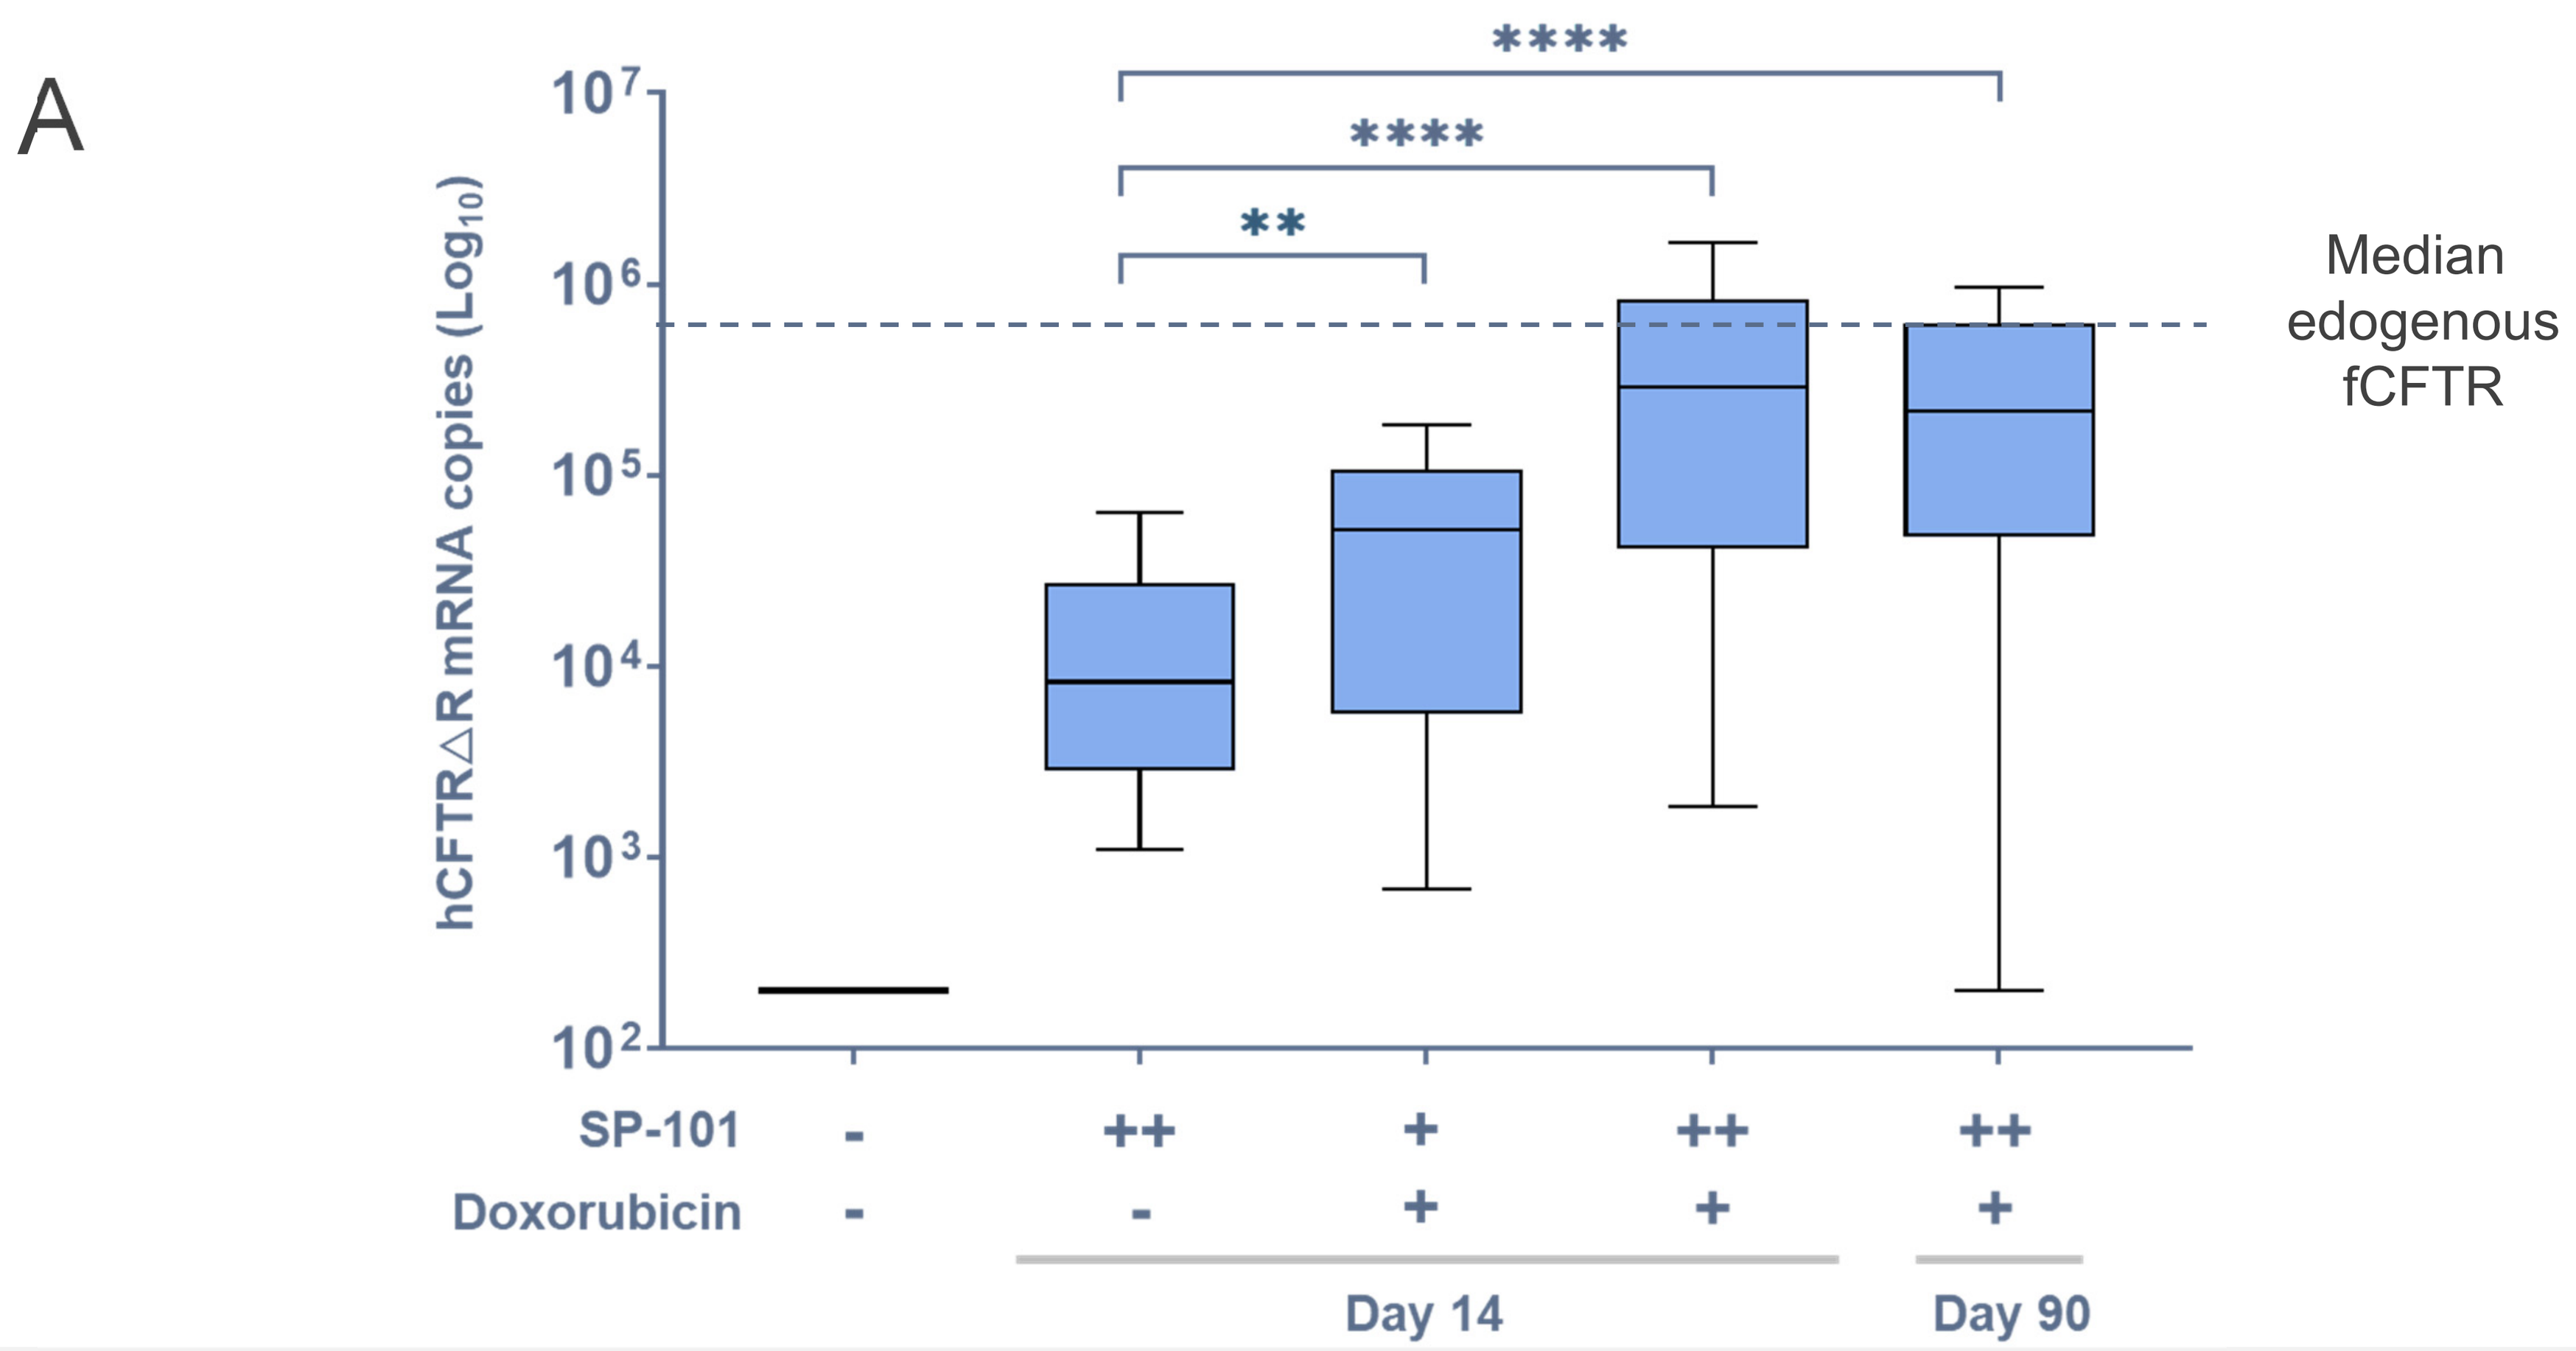
A

**
Median edogenous fCFTR
hCFTRΔR mRNA copies (Log10)
10^7
10^6
10^5
10^4
10^3
10^2
SP-101 - ++ + ++ ++
Doxorubicin - - + + +
Day 14
Day 90

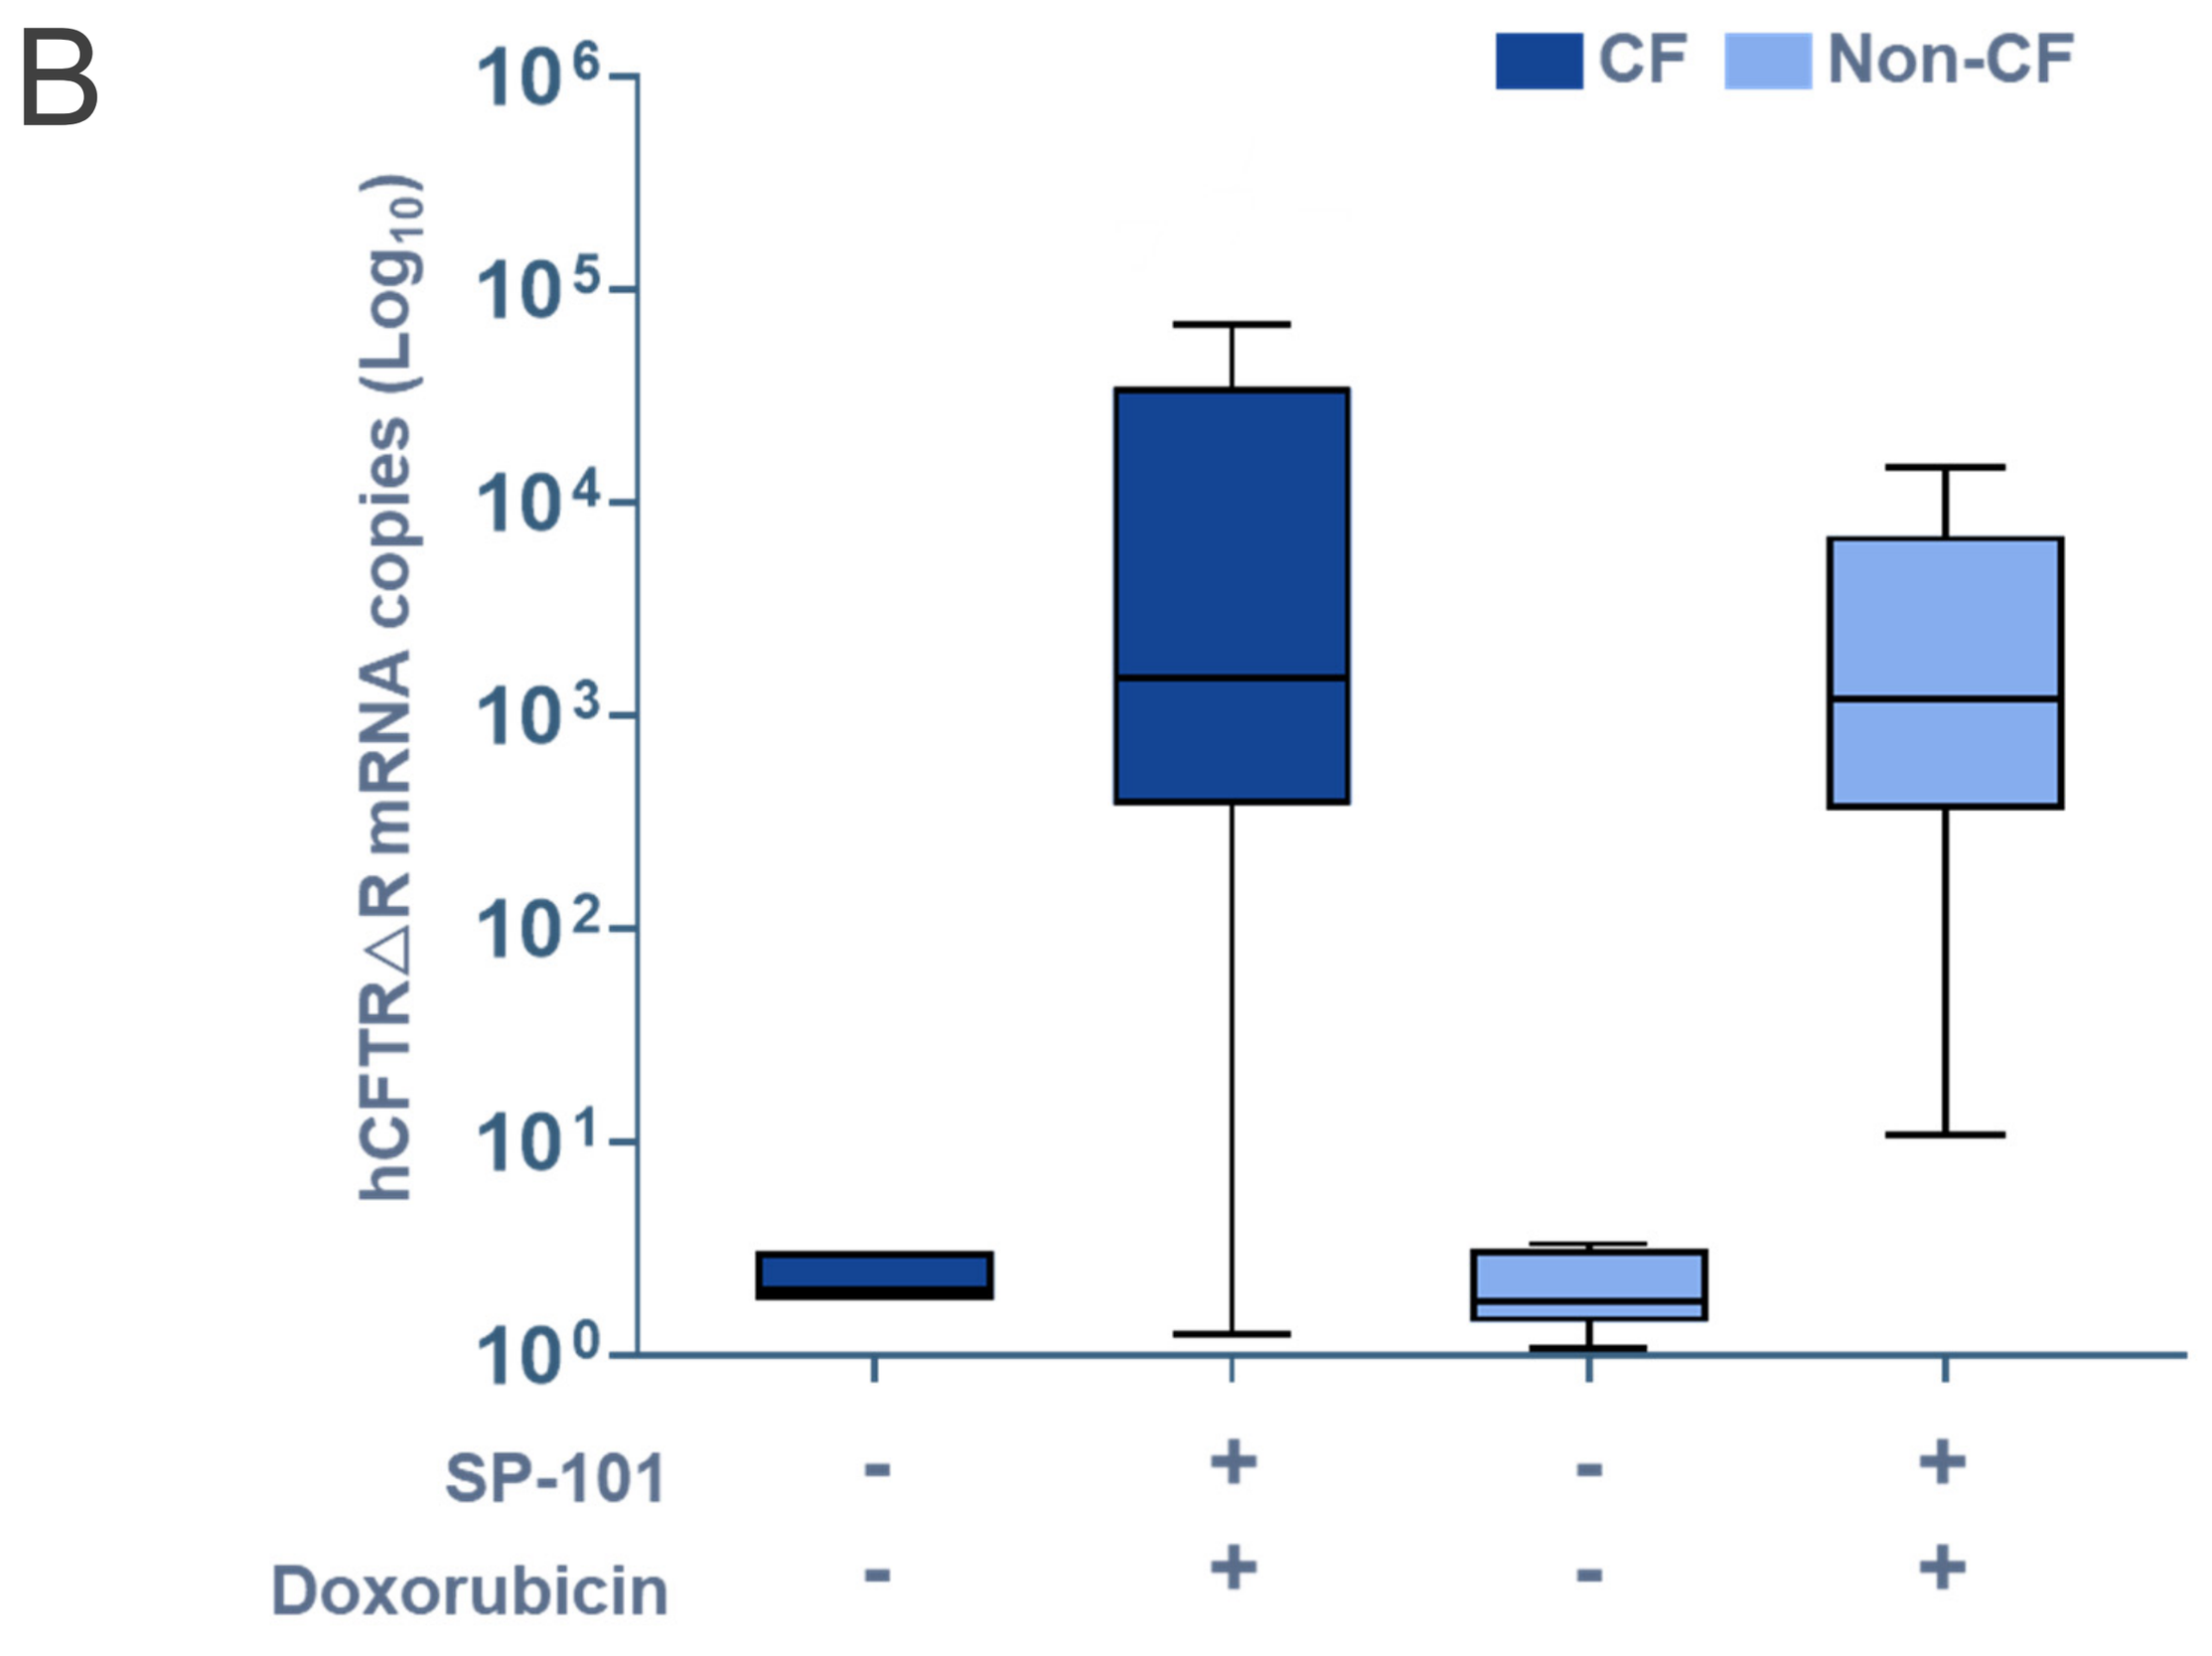
B
CF
Non-CF
hCFTRΔR mRNA copies (Log10)
10^6
10^5
10^4
10^3
10^2
10^1
10^0
SP-101 - + - +
Doxorubicin - + - +

A) *hCFTRΔR* mRNA expression is increased >10 fold by administration of doxorubicin and is durable in non-CF ferret lungs. In contrast to control samples, hCFTRΔR mRNA was detected in the majority of samples from animals exposed to SP-101 alone. However, hCFTRΔR mRNA was >10-fold higher in samples from animals exposed to the same amount of SP-101 followed by dox ($p<0.0001$). Moreover, hCFTRΔR mRNA did not significantly decrease (NS) 12 weeks (end of study) post-administration, indicating durable expression.
B) *hCFTRΔR* mRNA expression is similar in the lungs of CF and non-CF ferrets. In contrast to control animals (diluent only), hCFTRΔR mRNA was detectable to a similar extent in both CF (G551D) and non-CF animals suggesting that the CF lung is not an additional barrier to SP-101.

SP-101 holds great promise for people living with CF

- Doxorubicin is required for efficient CF correction
- SP-101 is tropic to many airway cell types
- hCFTRΔR mRNA expression is dose responsive, durable, and reach endogenous fCFTR levels
- hCFTRΔR mRNA expression is similar in CF and non-CF ferrets, suggesting that the CF airway is not an additional barrier to SP-101

spirovant

REFERENCES: 1. Excoffon KJ, et al. *Proc Natl Acad Sci U S A*. 2009;106(10):3865-3870. **2**. Ostedgaard LS, et al. *Proc Natl Acad Sci U S A*. 2002;99(5):3093-3098. **3**. Yan Z, et al. *Hum Gene Ther*. 2015;26(6):334-346. **4**.Yan Z, et al. *J Virol*. 2004;78(6):2863-2874. **5**. Zhang LN, et al. *Mol Ther*. 2004;10(6):990-1002. **6**. Tang Y, et al. *Mol Ther Methods Clin Dev*. 2020;19:186-200. **7**. Sun X, et al. *Sci Transl Med*. 2019;11(485):eaau7531.